Characterization of Gene Fusions in Solid Tumors Detected by RNA-Based Next-Generation Sequencing (NGS) Testing

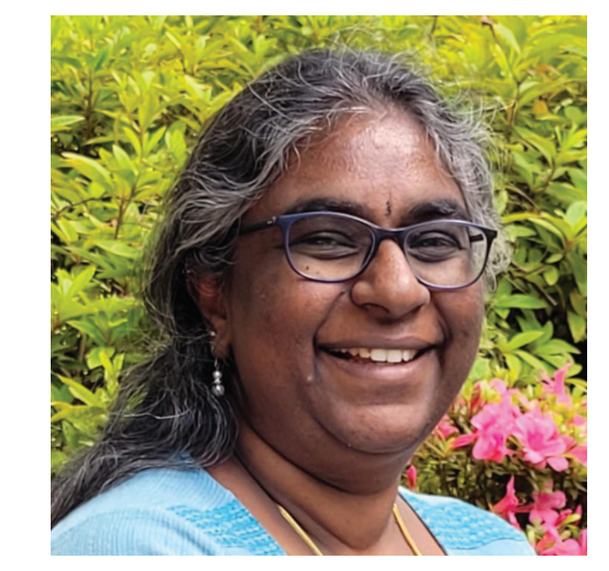

Contact

Aarthi Ravichandran
Director - Variant Science

VISIT LINKEDIN

+91 96860 31021

aarthi@strandls.com

strand

VISIT OUR **WEBSITE**

Aarthi Ravichandran[1], Bhanumathy G[1], Sreelaksh Raju[1], Swetha Nayanala[1], Kaushiki[1], Divya A. Priya[1], Srivathsan Adimoolam[1], Madhula Segaran[1], Mithua Ghosh[2], Sheela M. Lingaraju[2], Sameer Phalke[1], Shanmukh Katragadda[1], Vamsi Veeramachaneni[1], Urvashi Bahadur[1]

[1]Strand Life Sciences, Bengaluru, India

[2]Triesta Sciences (HealthCare Global Enterprises Ltd), Bengaluru, India

Background

Next-generation sequencing (NGS) based molecular testing has become the cornerstone of treatment management in advanced solid tumors. Larger NGS panels incorporate RNA-based gene fusion detection to help identify clinically relevant rearrangements. Gene fusions have therapeutic, diagnostic and prognostic value in solid tumors. To better understand the diversity in breakpoints and gene partners of gene fusions, we characterized gene fusions detected in solid tumor samples using 2 different panels, Illumina TSO500 and a custom 74-gene panel (SA74).

Methods

TSO 500 Panel
55 RNA genes
1953 samples

SA74 Panel
14 RNA genes
859 samples

RNA-based sequencing of 2812 FFPE samples

Analysis

Functional Significance evaluated considering the following

- Frame (only inframe fusions considered)
- Orientation (Effector gene 5′ or 3′)
- Gene Partner importance (only effector genes with gene partners that are functionally/biologically relevant are considered)

Results

Distribution of Fusion Positive Cases by Effector Gene

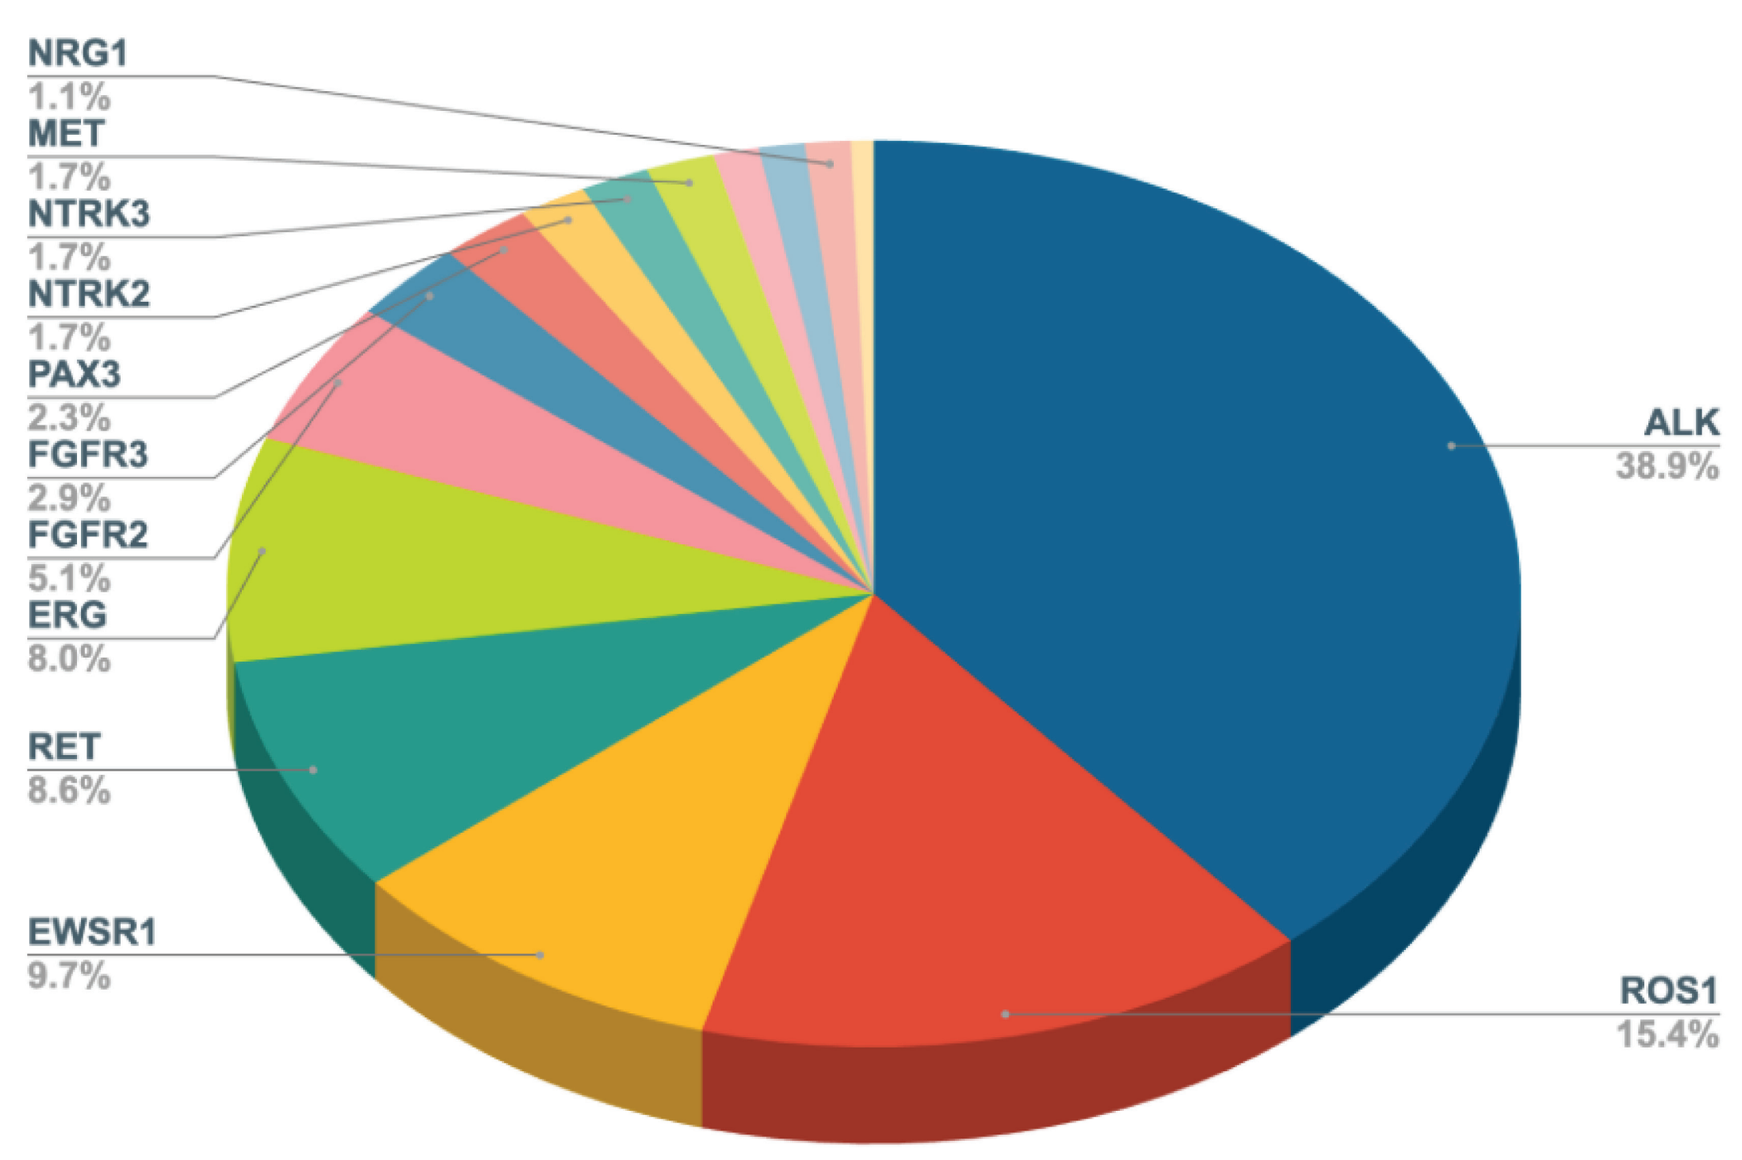

NRG1
1.1%
MET
1.7%
NTRK3
1.7%
NTRK2
1.7%
PAX3
2.3%
FGFR3
2.9%
FGFR2
5.1%
ERG
8.0%
RET
8.6%
EWSR1
9.7%
ALK
38.9%
ROS1
15.4%

Figure 1: Percentage of fusion positive cases by effector gene

Actionable (Tier1/2) gene fusions were detected in 175 cases across 15 effector genes. The most frequently altered genes were ALK (68/175;38.9%), ROS1 (27/175;15.4%), EWSR1 (17/175;9.7%), RET (15/175;8.6%) and ERG (14/175;8%).

Table 1: Distribution of fusion positive cases by effector gene

Gene	Case count	Percent cases
ALK	68	38.9%
ROS1	27	15.4%
EWSR1	71	9.7%
RET	15	8.6%
ERG	14	8.0%
FGFR2	9	5.1%
FGFR3	5	2.9%
PAX3	4	2.3%
NTRK2	3	1.7%
NTRK3	3	1.7%
MET	3	1.7%
BRAF	2	1.1%
NTRK1	2	1.1%
NRG1	2	1.1%
FGFR1	1	0.6%
Total	**175**	**100%**

Distribution of Fusion Positive Cases by Cancer Type

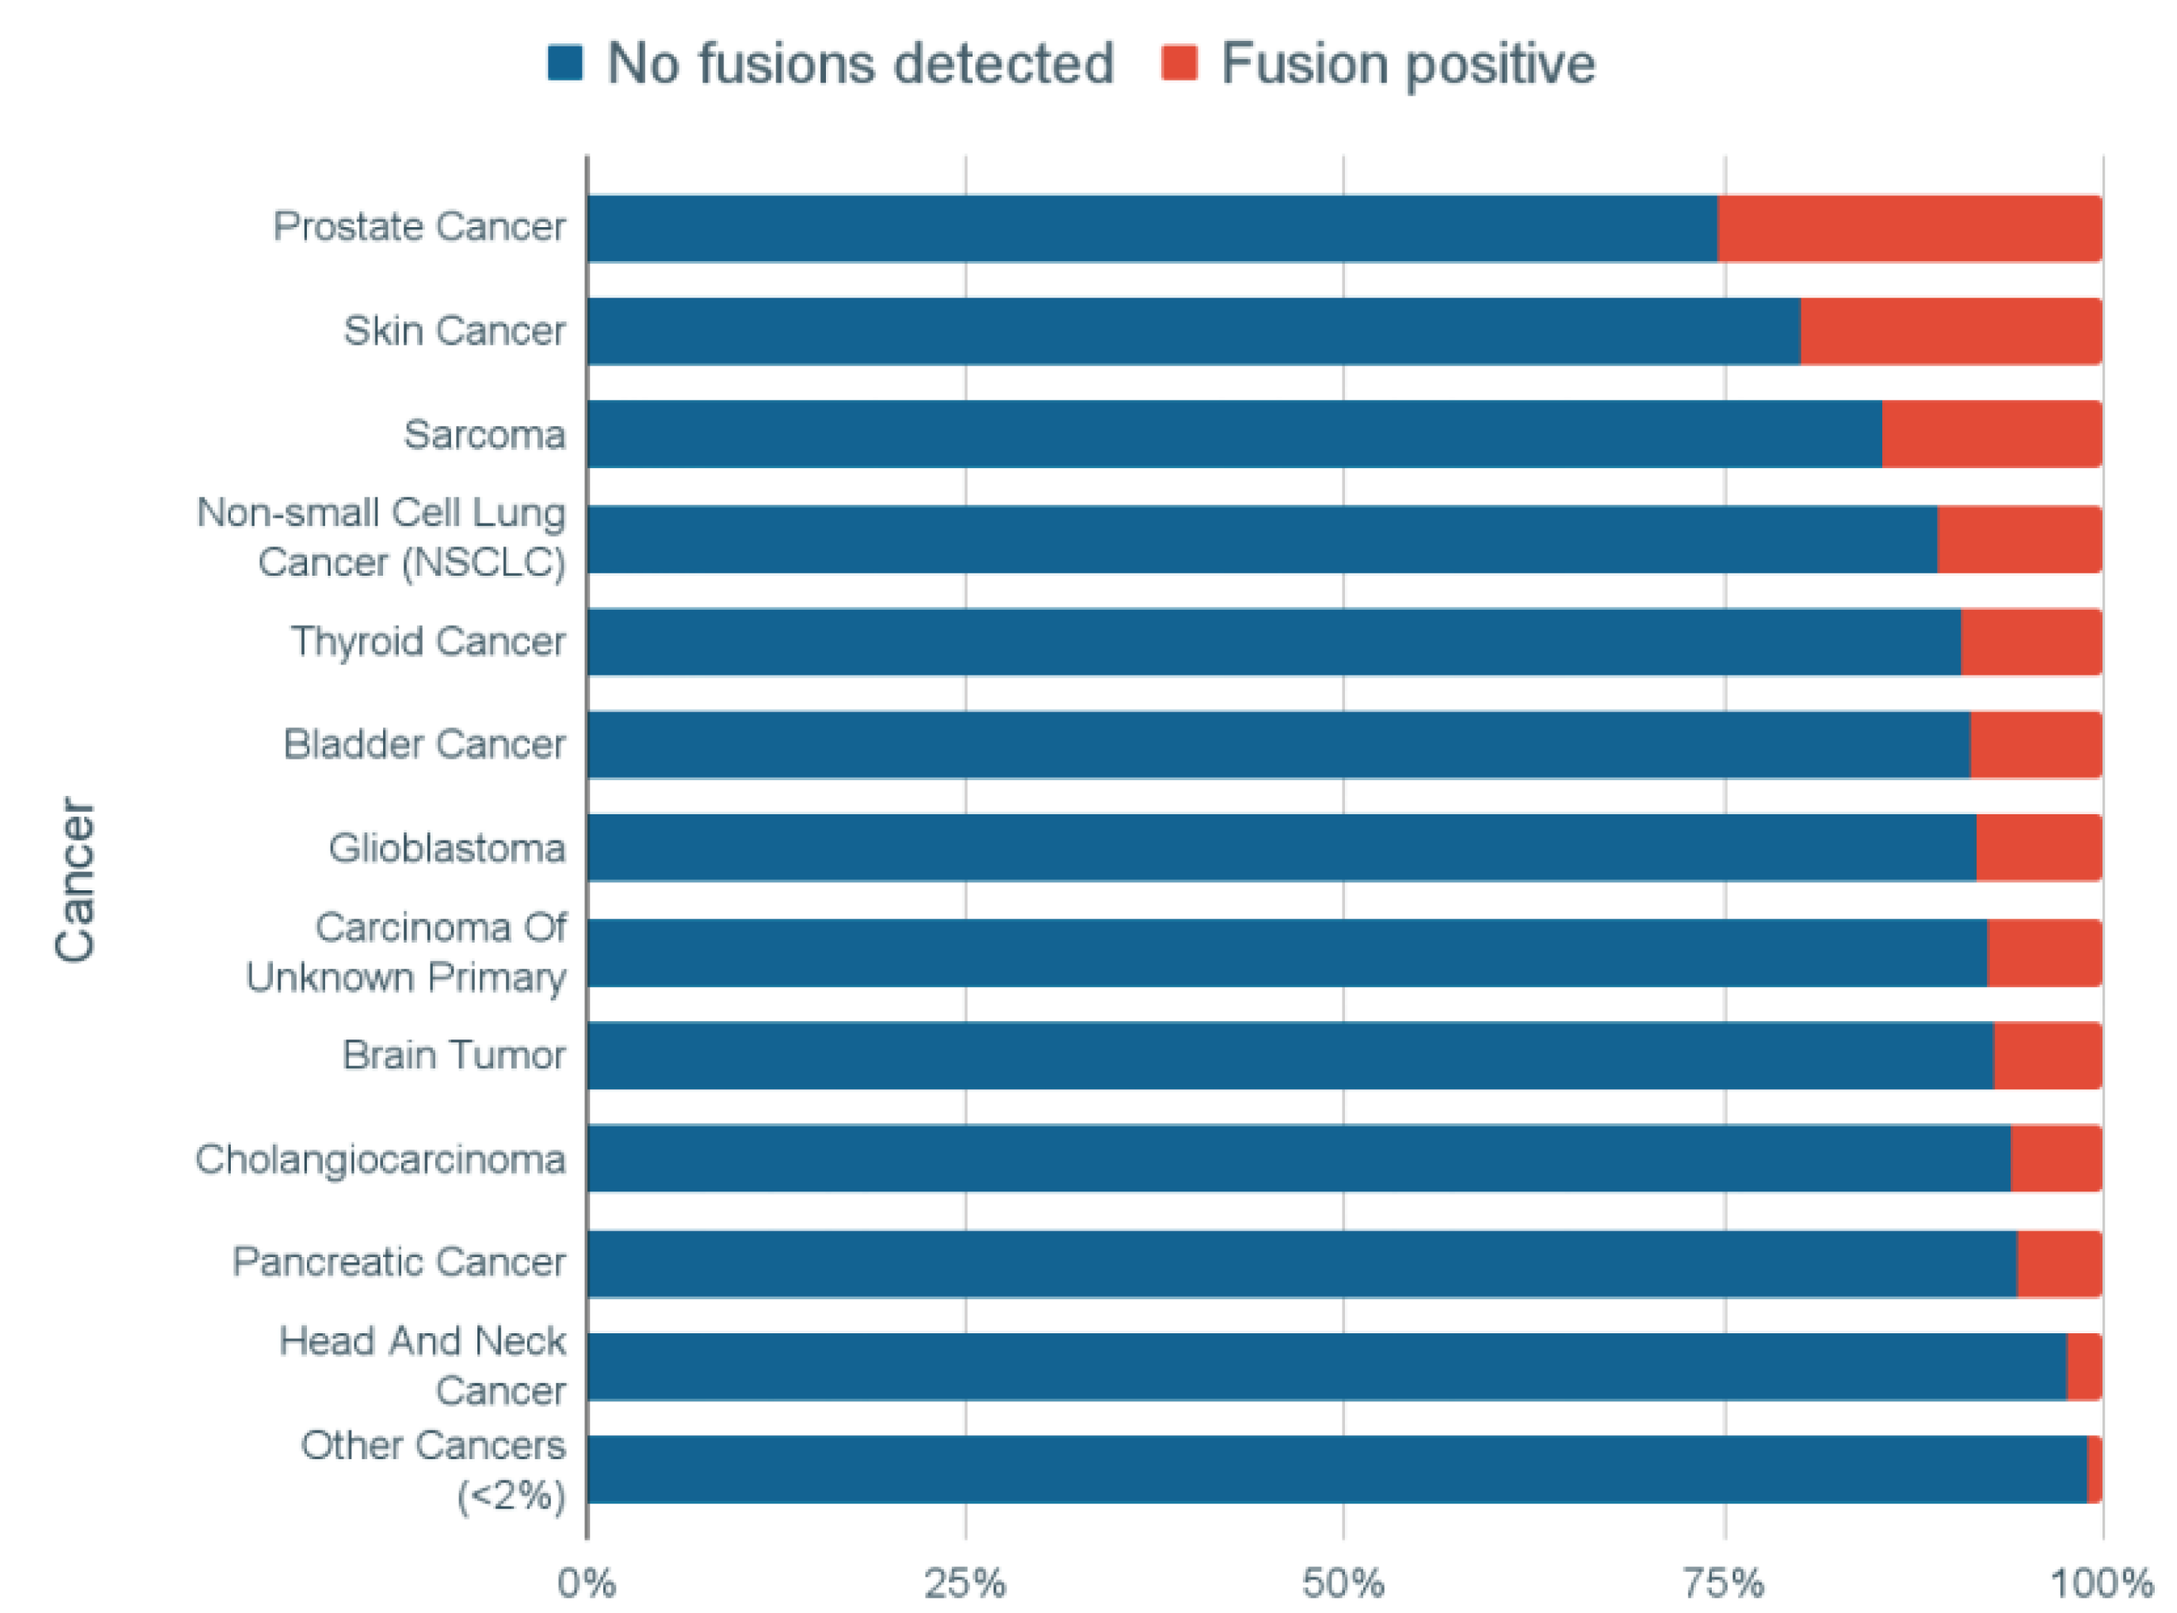

No fusions detected
Fusion positive
Cancer
Prostate Cancer
Skin Cancer
Sarcoma
Non-small Cell Lung Cancer (NSCLC)
Thyroid Cancer
Bladder Cancer
Glioblastoma
Carcinoma Of Unknown Primary
Brain Tumor
Cholangiocarcinoma
Pancreatic Cancer
Head And Neck Cancer
Other Cancers (<2%)
0%
25%
50%
75%
100%

Figure 2: Percentage of Fusion Positive Cases by Cancer Type

Table 2: Distribution of fusion positive cases by cancer type

Cancer Type	Total Cases	Fusion Positive	% Fusion Positive
Prostate Cancer	59	15	25.4%
Skin Cancer	5	1	20.0%
Sarcoma	137	20	14.6%
Non-small Cell Lung Cancer (NSCLC)	963	105	10.9%
Thyroid Cancer	54	5	9.3%
Bladder Cancer	23	2	8.7%
Glioblastoma	12	1	8.3%
Carcinoma Of Unknown Primary	106	8	7.5%
Brain Tumor	14	1	7.1%
Cholangiocarcinoma	83	5	6.0%
Pancreatic Cancer	18	1	5.6%
Head And Neck Cancer	178	4	2.2%
Other Cancers (<2%)	707	7	1.0%
Gastric (stomach) Cancer	61	1	1.6%

Gene fusions were detected across cancers; prostate cancer(15/59;25.4%), sarcoma (20/137;14.6%), non-small cell lung cancer (NSCLC)(105/963;10.9%), thyroid cancer (5/54;9.3%) and cholangiocarcinoma (5/83;6.0%) had the highest frequencies.

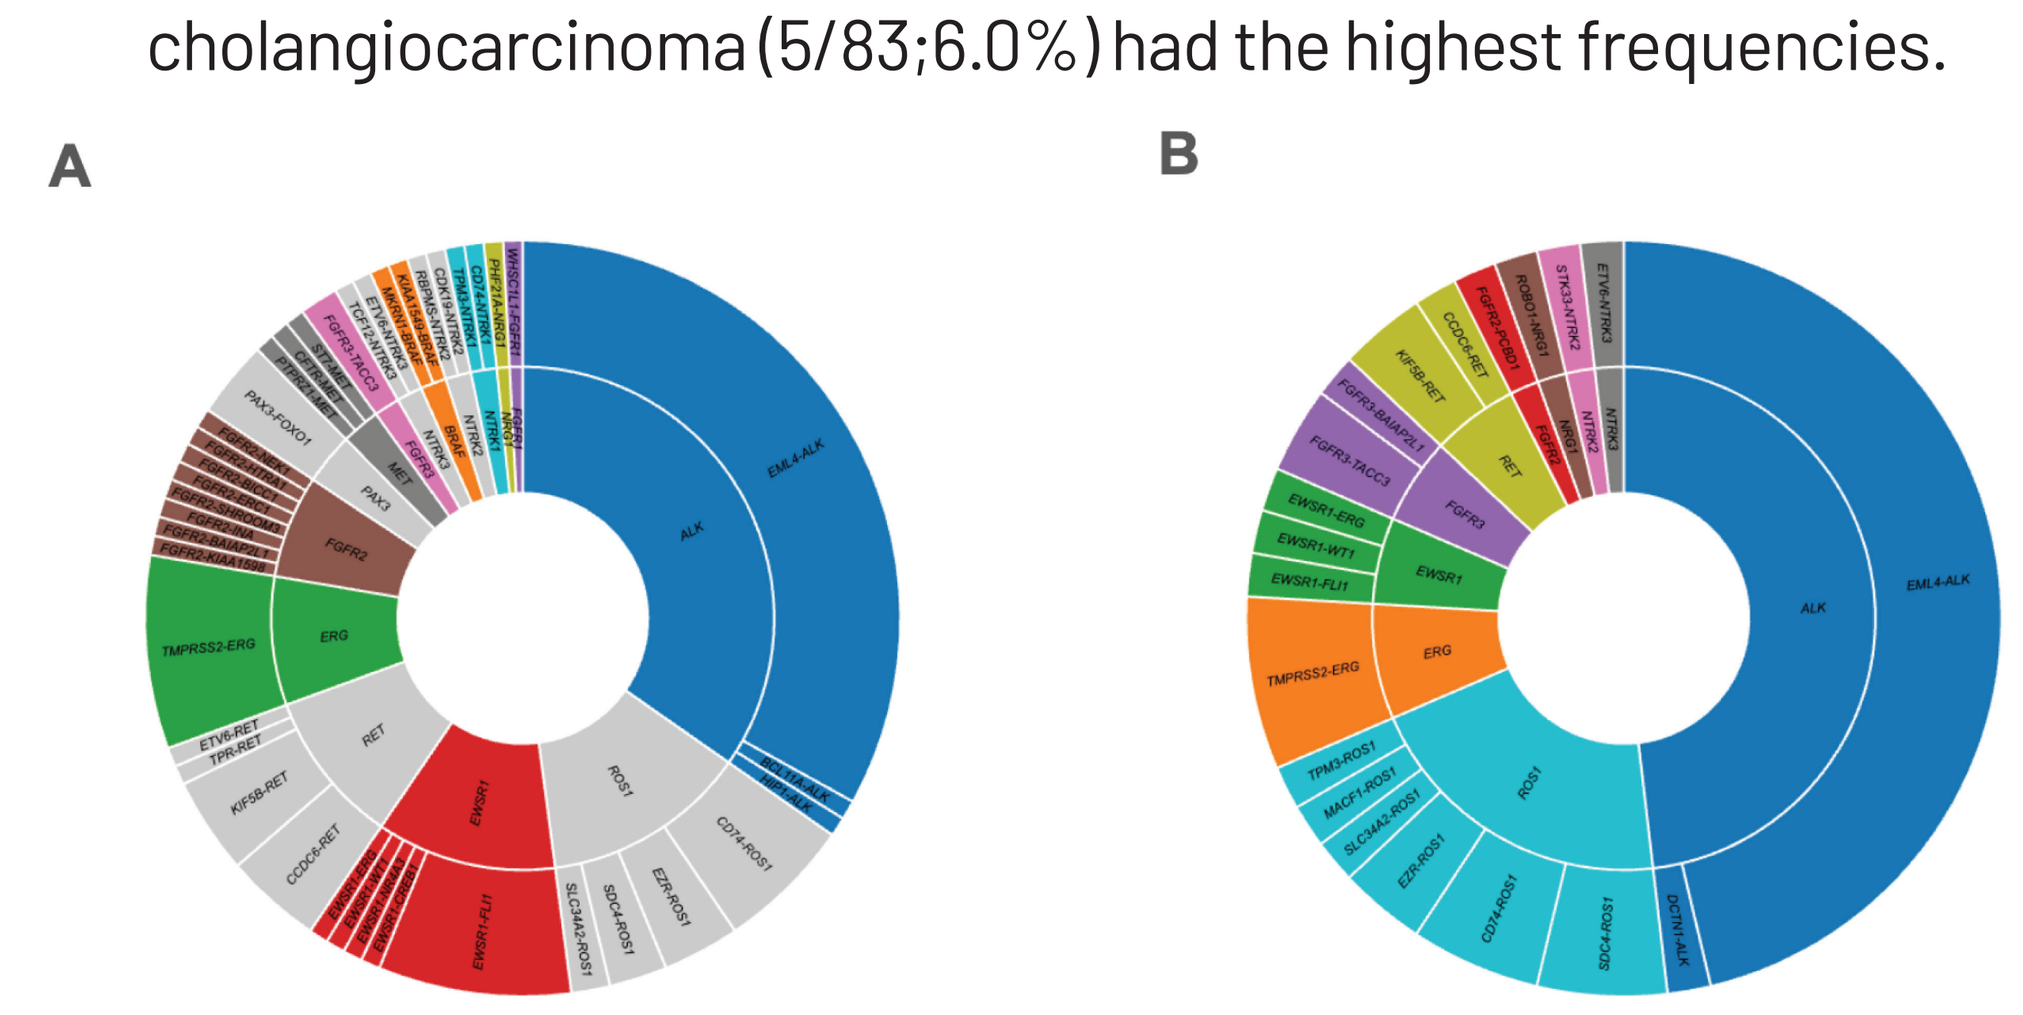

A
B

Figure 3: Fusion variants by effector gene detected on the (A) TSO500 and (B) SA74 panels

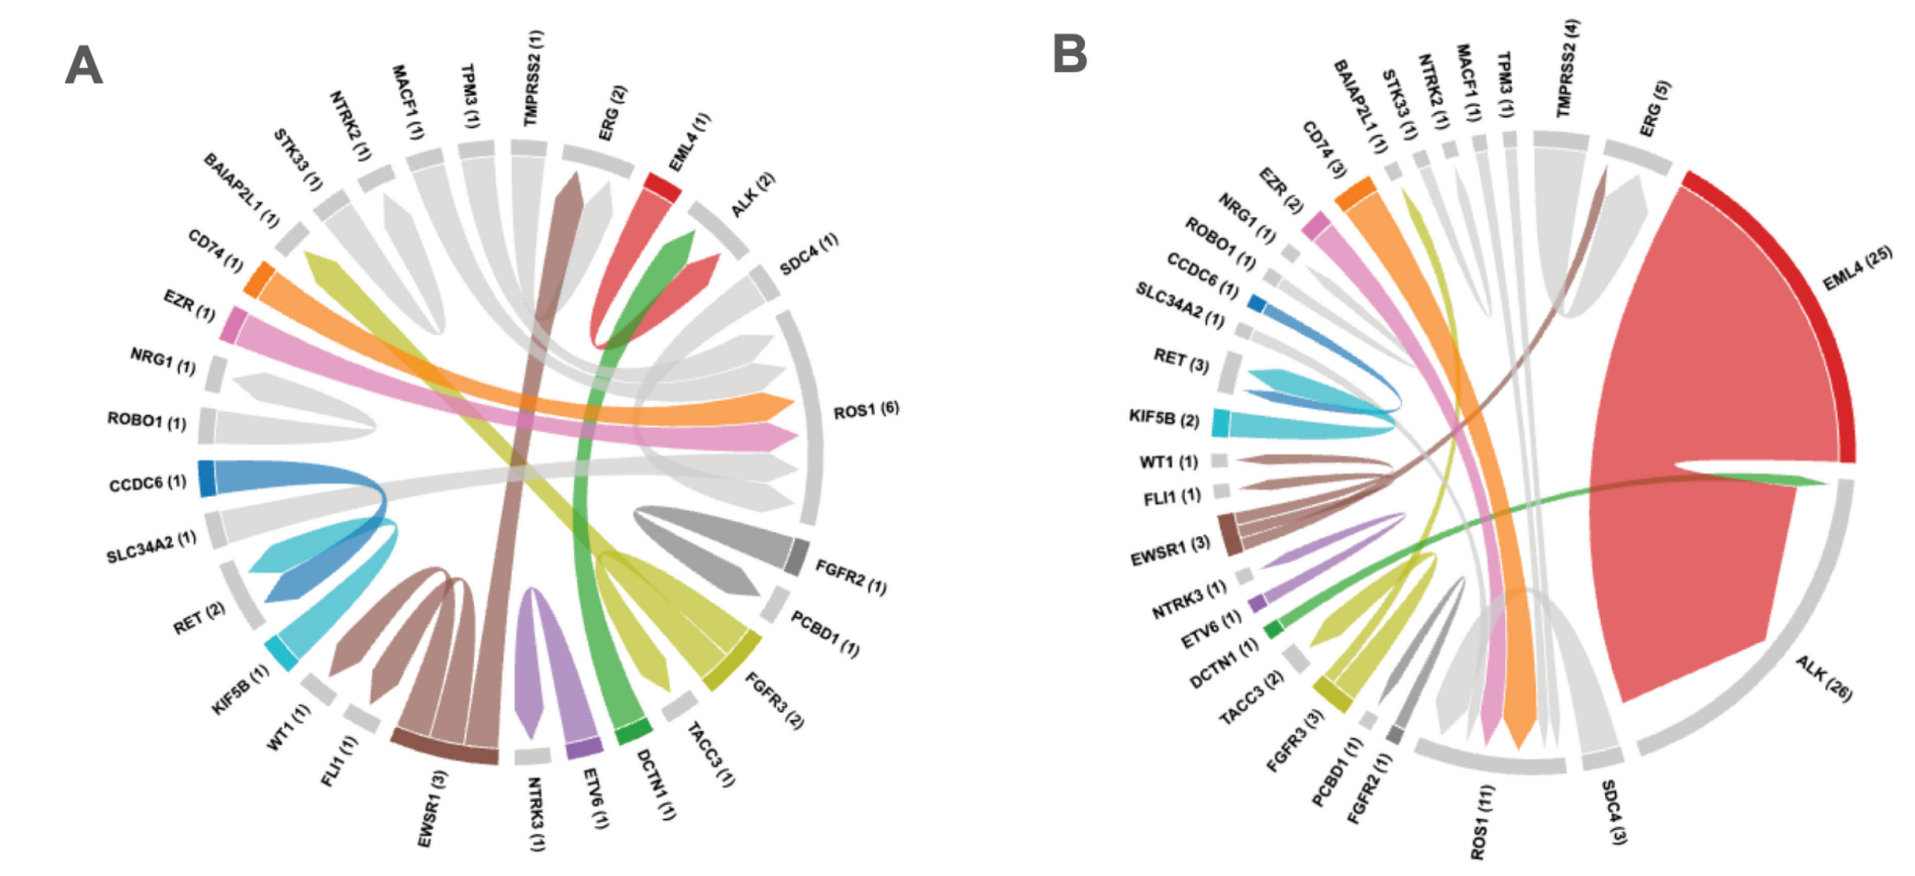

A
B

Figure 4: Distribution of the fusions detected on the SA74 panel with (A) number of partner genes and (B) case count per effector gene

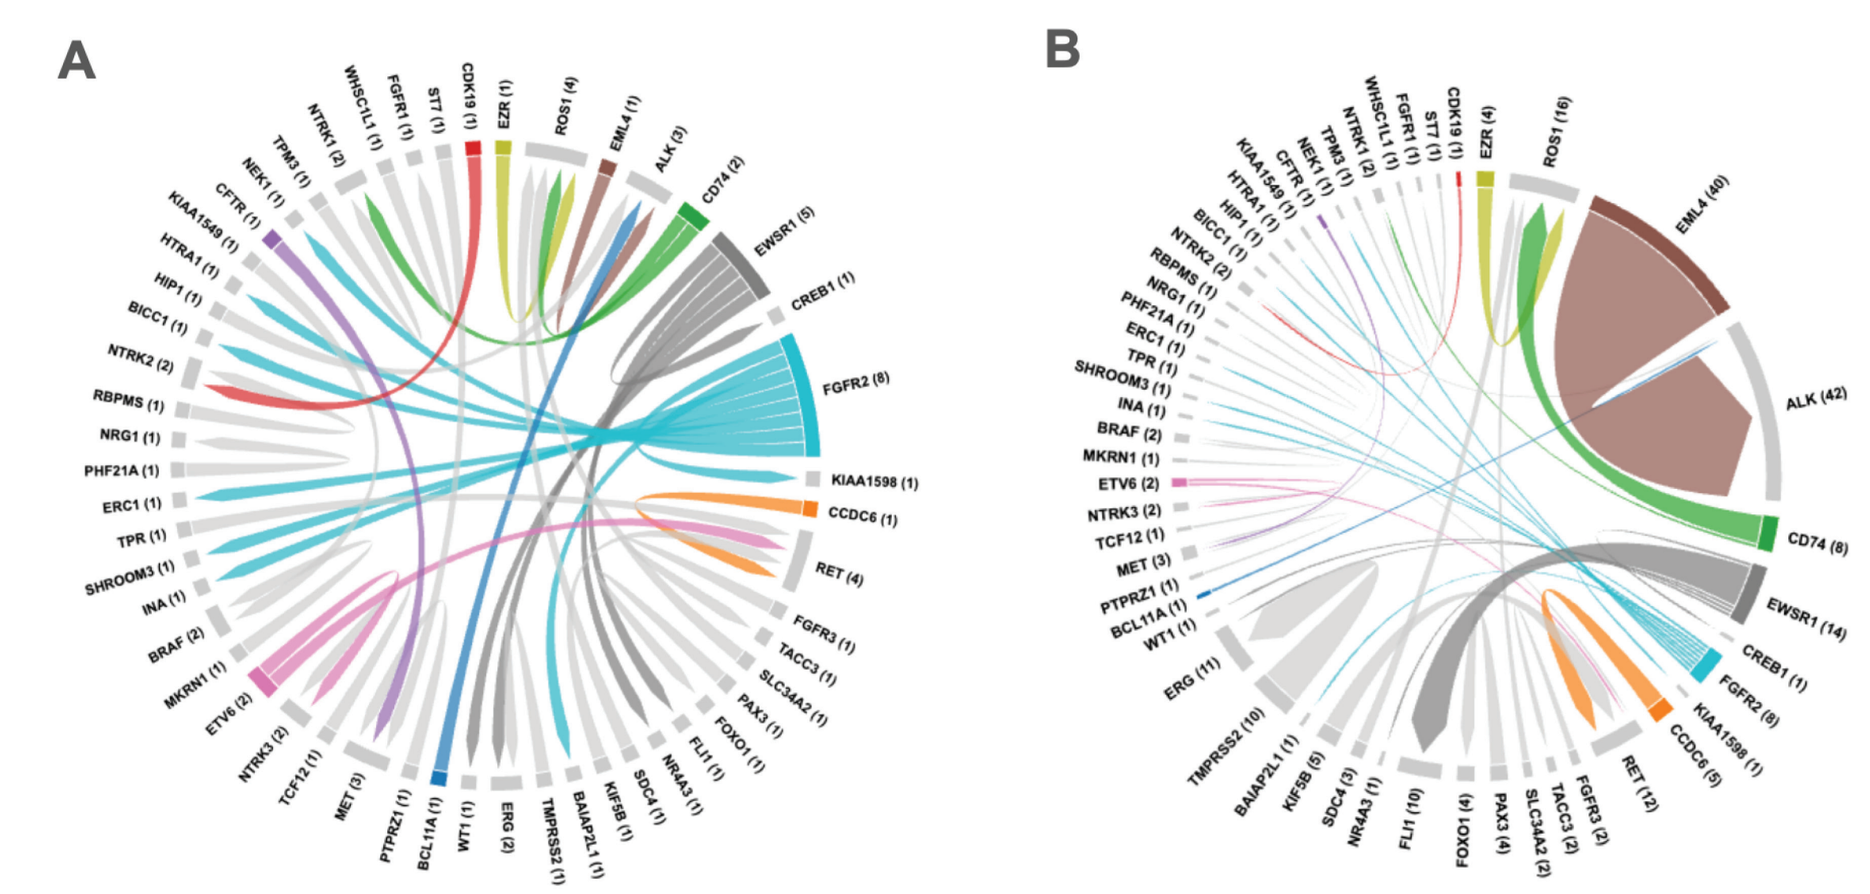

A
B

Figure 5: Distribution of the fusions detected on the TSO500 panel with (A) number of partner genes and (B) case count per effector gene

Gene Partners and Breakpoints are Important for Gene Fusion Characterization

Gene fusions are characterised using gene partners and exon breakpoints. Gene partners often provide either the interaction domains or promoter regions in cases where the active partner expression is increased in a fusion protein.

- Most of the ALK gene fusions had EML4 (65/68) as the partner gene
- Some genes had multiple different partners such as FGFR2 (9 partners), ROS1 (6 partners), EWSR1 (5 partners)
- ERG was observed with only one partner, TMPRSS2
- Although most fusion partners were well known, we observed novel partners in a few cases (FGFR2-HTRA1, FGFR2-PCBD1, MACF1-ROS1)

Diversity in Exon Breakpoints Observed in Certain Genes

Gene fusion proteins may have the same or different breakpoints. The breakpoints define the functional region of the protein required for a relevant fusion product to be formed.

- All ALK gene fusions had a breakpoint at exon 20 of ALK
- RET gene fusions had a breakpoint at exon 12 of RET
- FGFR2/3 gene fusions had a breakpoint at exon 17 of FGFR2/3
- However, for ROS1 multiple breakpoints were observed, mainly at exon 32/34, with other exons observed at lower frequencies
- EWSR1 also showed multiple breakpoints with the majority of them having a breakpoint at exon 7

Conclusion

RNA-based NGS testing allows for detection of multiple different gene fusion partners. This diversity is relevant for multiple genes such as FGFR2/3 and NTRK1/2/3. Further, specific breakpoint determination is critical for determining the clinical significance of the fusion proteins. Gene fusions, while rare, are a key component of comprehensive genomic profiling of solid tumors. RNA-based NGS therefore offers a superior method for gene fusion evaluation.

Highlights

- **RNA-based** gene fusion detection is a common feature of **large NGS panels**, aiming to incorporate detection of **clinically relevant rearrangements**.
- Gene fusions detected on two panels, **TSO500** and **SA74** were characterized.
- RNA-based testing allows for detection of **multiple different fusion partners** for the same effector gene and also covers **multiple different exon breakpoints** for the same effector gene.
- This work thus demonstrates that **RNA-based fusion detection**, as a component of comprehensive genomic profiling, provides **high utility** as it detects a **diverse array** of clinically relevant gene fusions.

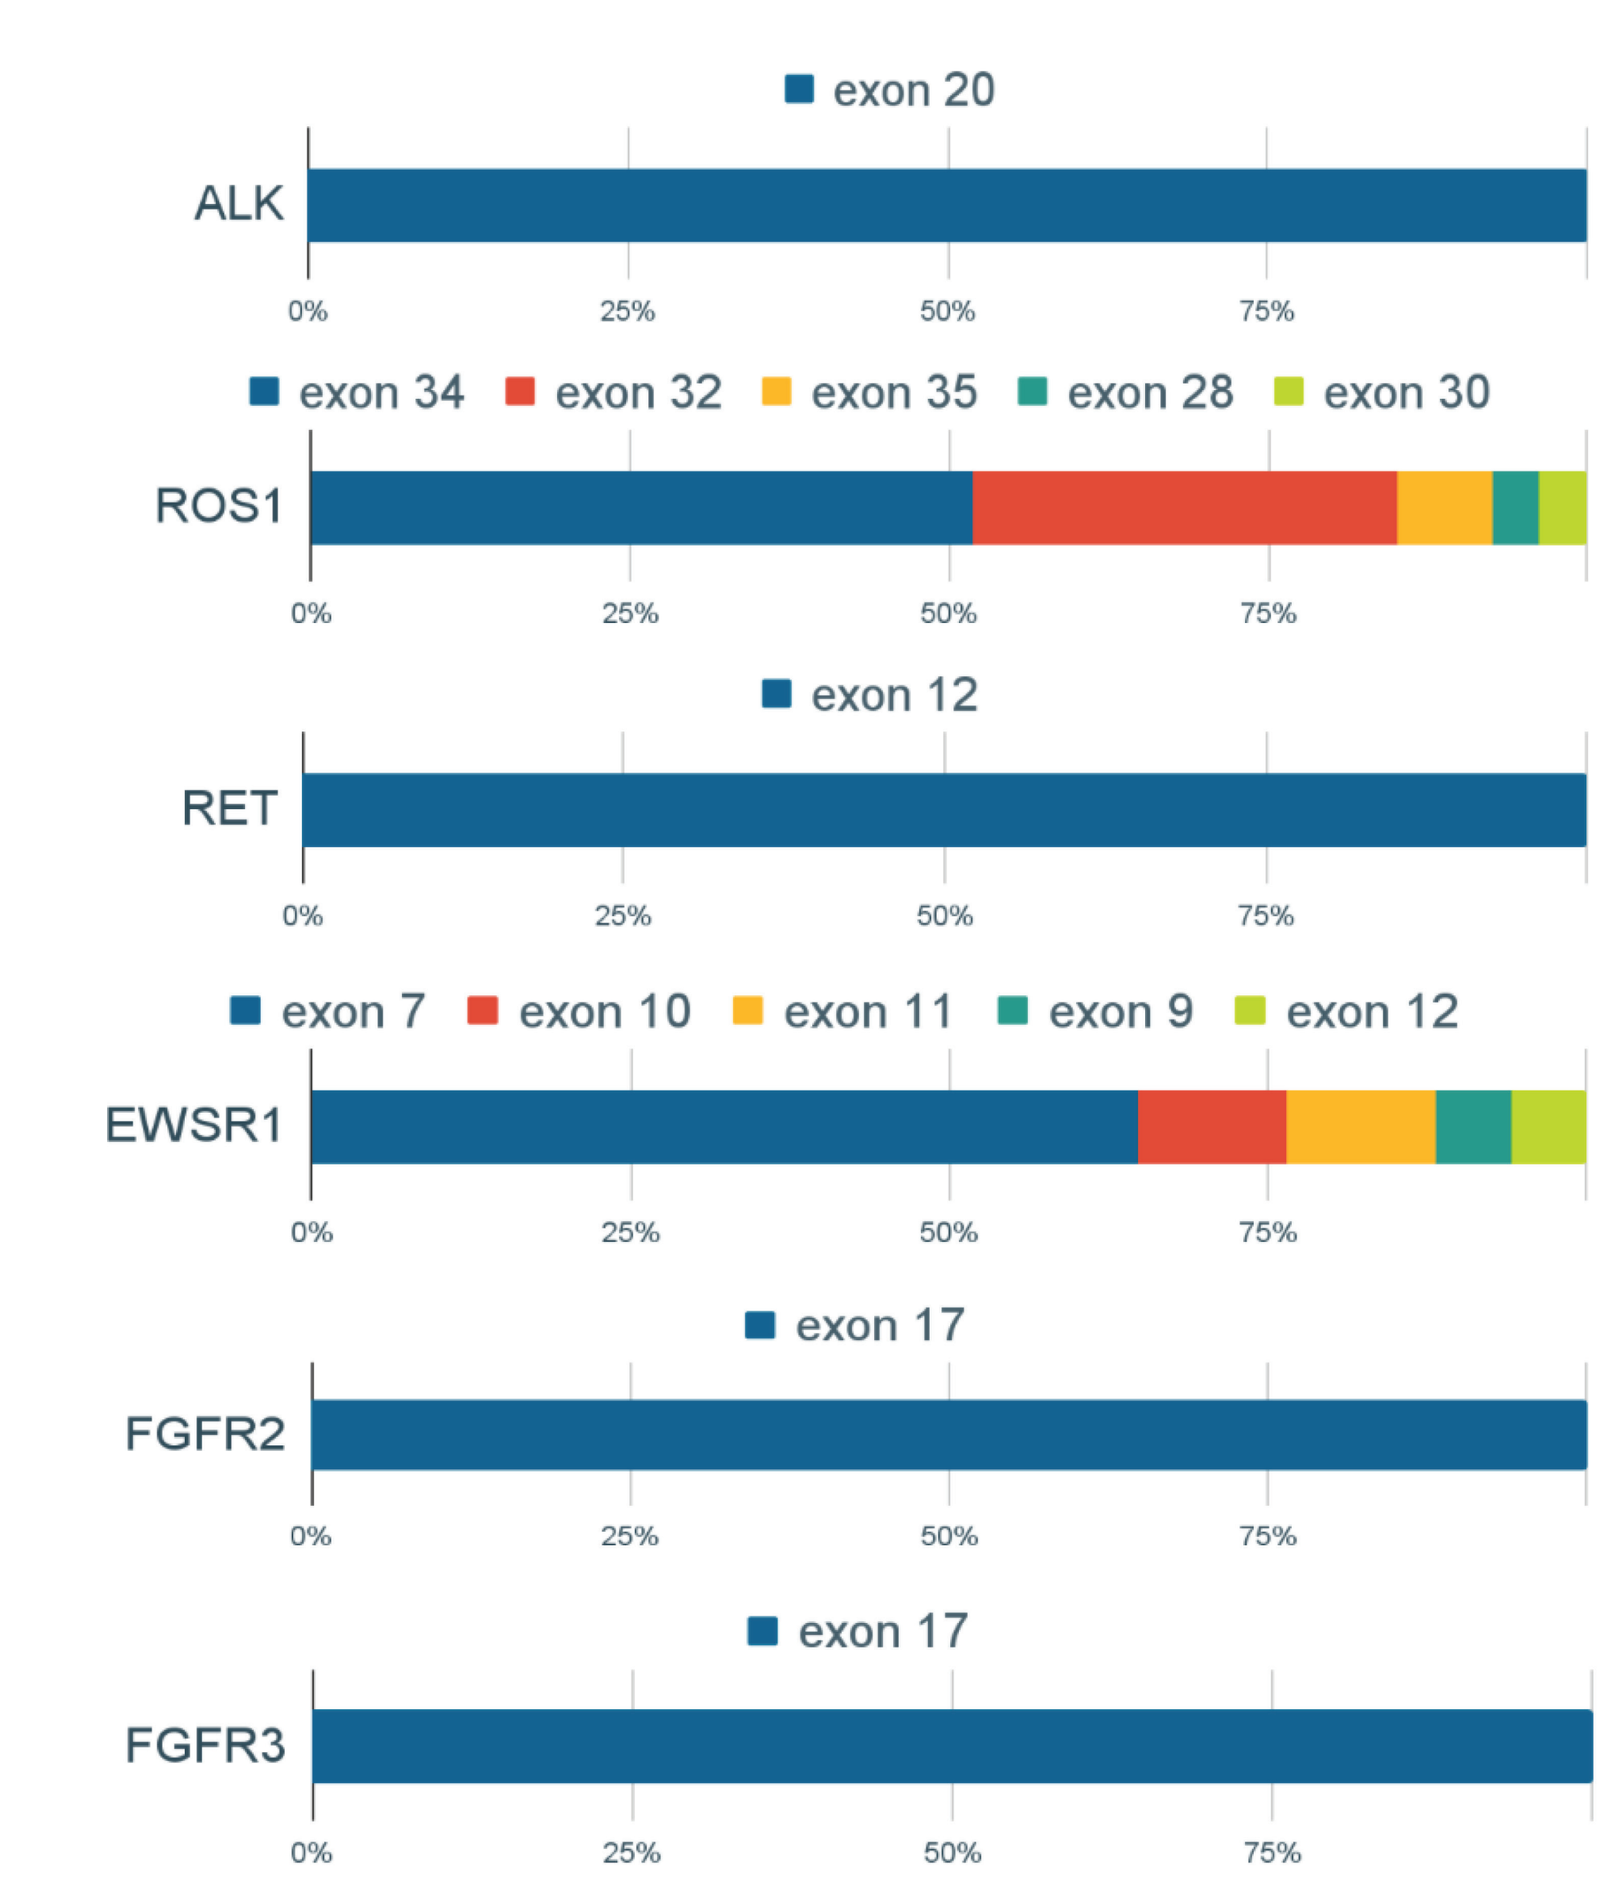

exon 20
ALK
exon 34
exon 32
exon 35
exon 28
exon 30
ROS1
exon 12
RET
exon 7
exon 10
exon 11
exon 9
exon 12
EWSR1
exon 17
FGFR2
exon 17
FGFR3
0%
25%
50%
75%

Figure 6: Breakpoint distribution for different gene fusion effector proteins